Altasciences, 6605 Merrill Creek Pkwy, Everett, WA 98203
altasciences.com | contact@altasciences.com

Assessment of Receptor Occupancy via Flow Cytometry: Benefits and Pitfalls of Two Common Approaches

Andrea S.L. Wong, Milan Patel, Floyd Wagner, Martin Poirier
Altasciences, Seattle, WA, United States

ABSTRACT

Receptor occupancy assays can be a powerful tool in the determination of drug dosage. Recently, flow cytometry has been the method of choice to assess receptor occupancy due to its ability to measure drug-target binding interactions on multiple cell populations simultaneously. Receptor occupancy assays using flow cytometry involve measurement of target receptors bound by the drug, number of receptors not bound to the drug, and the total number of receptors present. The methods for measuring these parameters, particularly for antibody-based therapeutics, can be categorized into two approaches based on the detection antibodies used. Here, we discuss the benefits and pitfalls of these methods.

In one approach, fluorescently labelled competing and non-competing antibodies to the drug are used to detect Free and Total Receptors, respectively. In this approach, the competing antibody is often the labelled drug itself. One of the main obstacles in method development is to identify a non-competing antibody with comparable binding affinity to the target as the drug tested. We have observed that when competing and non-competing antibodies display different affinities to the target, the results can be difficult to interpret. In addition, it is imperative that in a multiplex assay, competing and non-competing antibodies do not interfere with each other's binding to the target.

The other approach utilizes a single, fluorescently labelled secondary antibody to detect both Bound and Total Receptors. Samples from animals treated *in vivo* are first incubated with excess drug *ex vivo,* to fully saturate all targets, and stained with secondary antibody to detect total receptors. Staining with a single detection antibody results in a direct comparison between bound and total receptors, removing any difficulties from different binding affinities between reagents. We have observed that identifying a suitable secondary antibody is crucial, especially when the targeted populations consist of sticky cells, such as granulocytes and monocytes. In addition, we have found that incubation of samples *ex vivo* with excess drug can lead to down regulation of the target, leading to underestimation of the Total Receptors and overestimation of receptor occupancy of the drug.

The technical considerations presented here, when incorporated early in the receptor occupancy design phase, will aid in determining the most suitable approach, and avoid common pitfalls in assay design.

INTRODUCTION

Flow cytometry has long been a preferred technique used by immunologists to immunophenotype various immune cell populations and understand a plethora of cell functions, including proliferation, activation, and intracellular cytokine productions. With the breakthrough of immunotherapies as a new class of drug to treat cancers and other diseases, flow cytometry has emerged as the preferred method of evaluating receptor occupancy (RO) by monoclonal antibodies that target immune checkpoints or modulators. The failure of TGN1412 in its first-in-human clinical trials has further emphasized the need to assess RO, as a significantly lower dose of TGN1412 would have been recommended in human trials had RO assessment been performed during preclinical safety assesment. Assessment of RO by these monoclonal antibodies at the preclinical phase not only provides invaluable information with regards to dosage in clinical trials, it also demonstrates that the relevant species has been selected for safety assessment through adequate target binding.

Assessment of RO by test article (TA) using flow cytometry involves the measurement of target receptors bound by the TA (Bound Receptor), number of receptors not bound to the TA (Free Receptor), and the total number of receptors present (Total Receptor). The methods to assess RO can also be categorized based on the detection reagents used such as 1) antibodies that compete and do not compete with the TA and 2) anti-TA secondary human IgG antibodies.

METHOD

Figures 1. RO assay with competing and non-competing antibodies

1a.

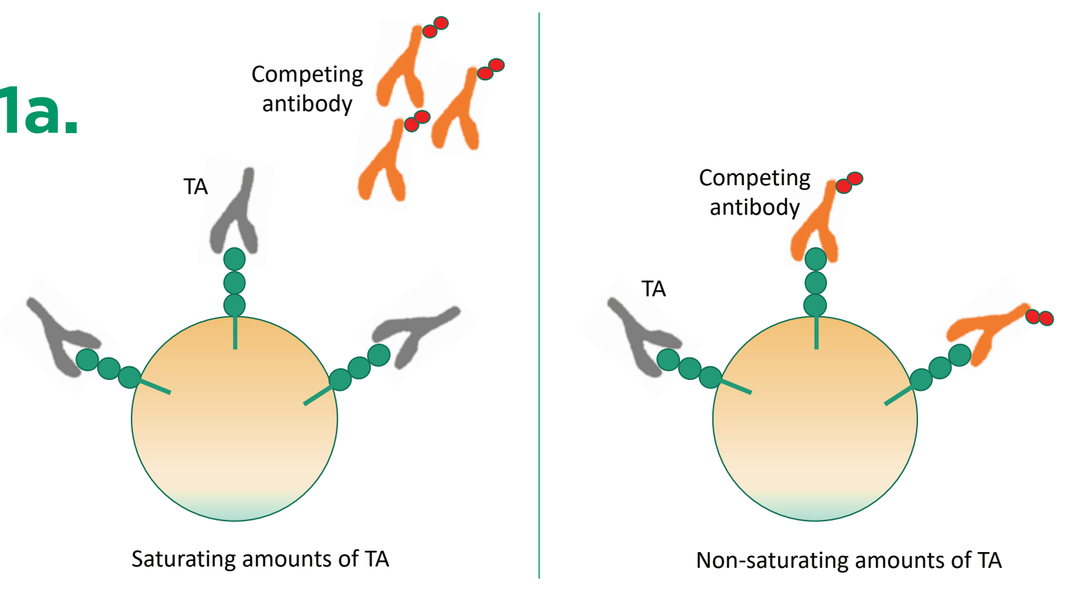
Competing antibody
TA
Saturating amounts of TA
Non-saturating amounts of TA

1b.

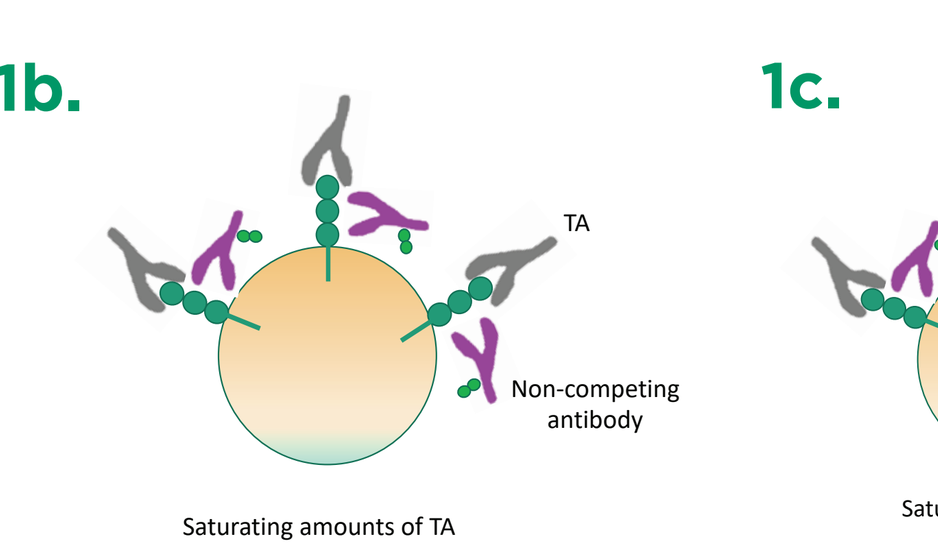
TA
Non-competing antibody
Saturating amounts of TA

1c.

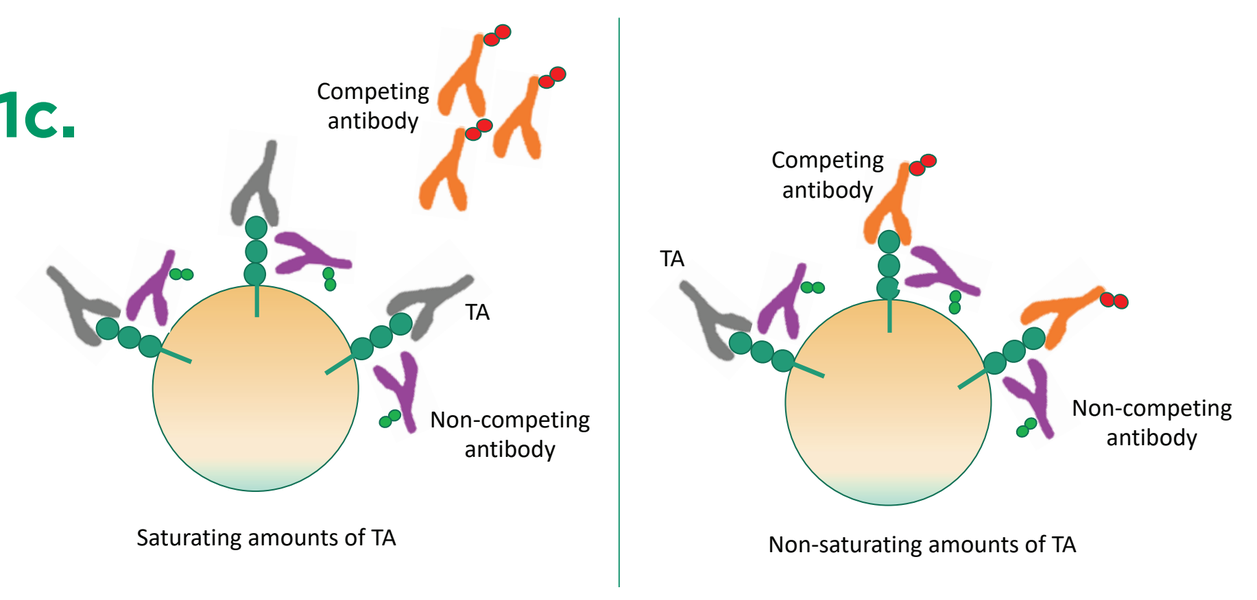
Competing antibody
TA
Non-competing antibody
Saturating amounts of TA
Non-saturating amounts of TA

Competing antibody measures Free Receptor and can either be a fluorescently labelled test article or another fluorescently labeled antibody known to compete with the TA for the therapeutic target. Under saturating amounts of TA, the competing antibody will have little to no binding to the therapeutic target. In contrast, under non-saturating amounts of TA, the competing antibody will bind to the therapeutic target and, therefore, report on the level of Free Receptor.

Non-competing antibody measures Total Receptor and is a fluorescently labelled antibody that binds to the therapeutic target, and does not interfere with the bind of TA or the competing antibody to the therapeutic target. Non-competing antibody is used to monitor Total Receptor expression throughout the study across multiple time points and enables the assessment of receptor modulation in response to TA administration.

RO assay with competing and non-competing antibodies. Samples for animals dosed with TA were co-stained with competing and non-competing antibodies to determine levels of Free and Total Receptor. Under saturating amounts of TA, only the non-competing antibody will bind to the therapeutic target while the competing antibody will not. As TA concentration decreases, the Free Receptor or therapeutic target becomes available and binding of the competing antibody is observed.

Figures 2. RO assay with single fluorescently labelled secondary antibody to detect both Bound and Total Receptors.

2a.

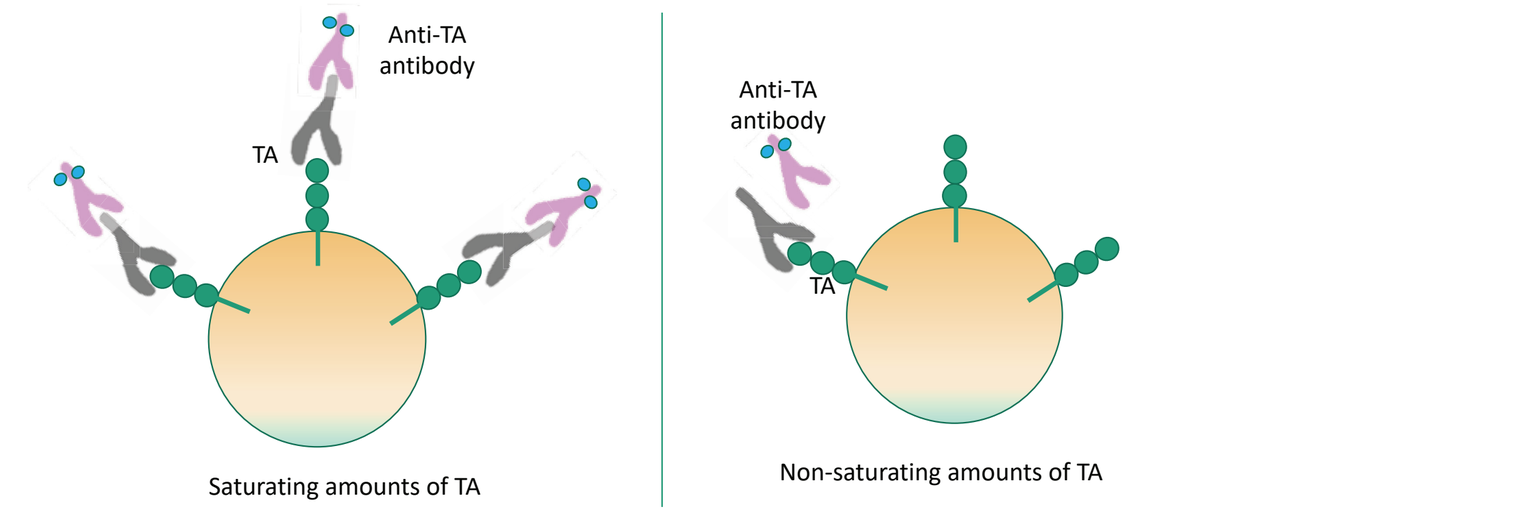
Anti-TA antibody
TA
Saturating amounts of TA
Non-saturating amounts of TA

Use of secondary antibody to measure Bound Receptor. Commercially available anti-human IgG antibodies are the commonly used secondary antibody to detect TA bound on the therapeutic target.

2b.

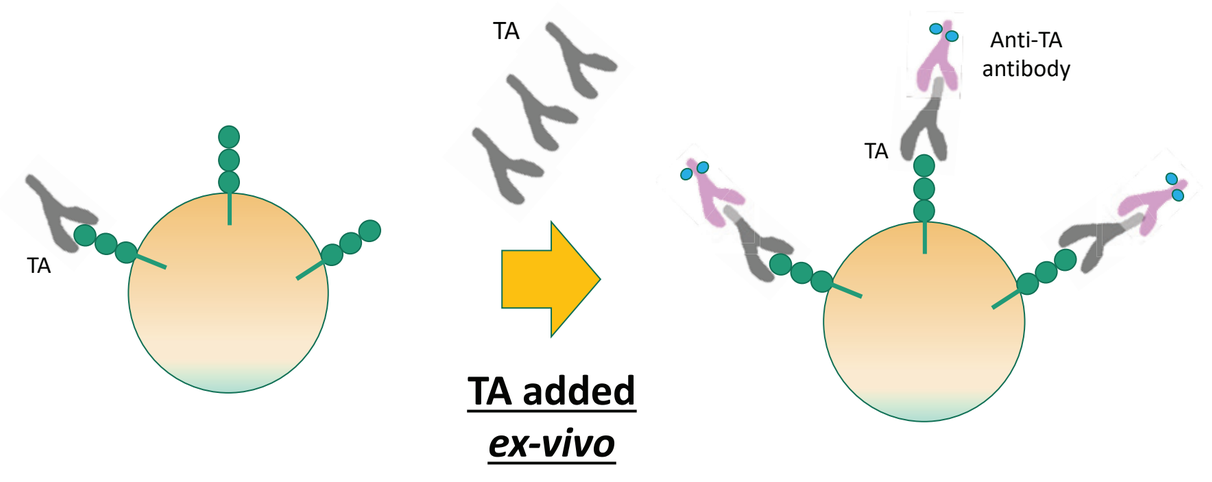
TA
Anti-TA antibody
TA added
ex-vivo

Use of secondary antibody to measure Total Receptor. The same anti-human IgG antibody used to detect Bound Receptor can be used to measure Total Receptor. To achieve this, samples will be incubated with TA *ex vivo* to saturate all available therapeutic targets, followed by staining with the secondary antibody.

2c.

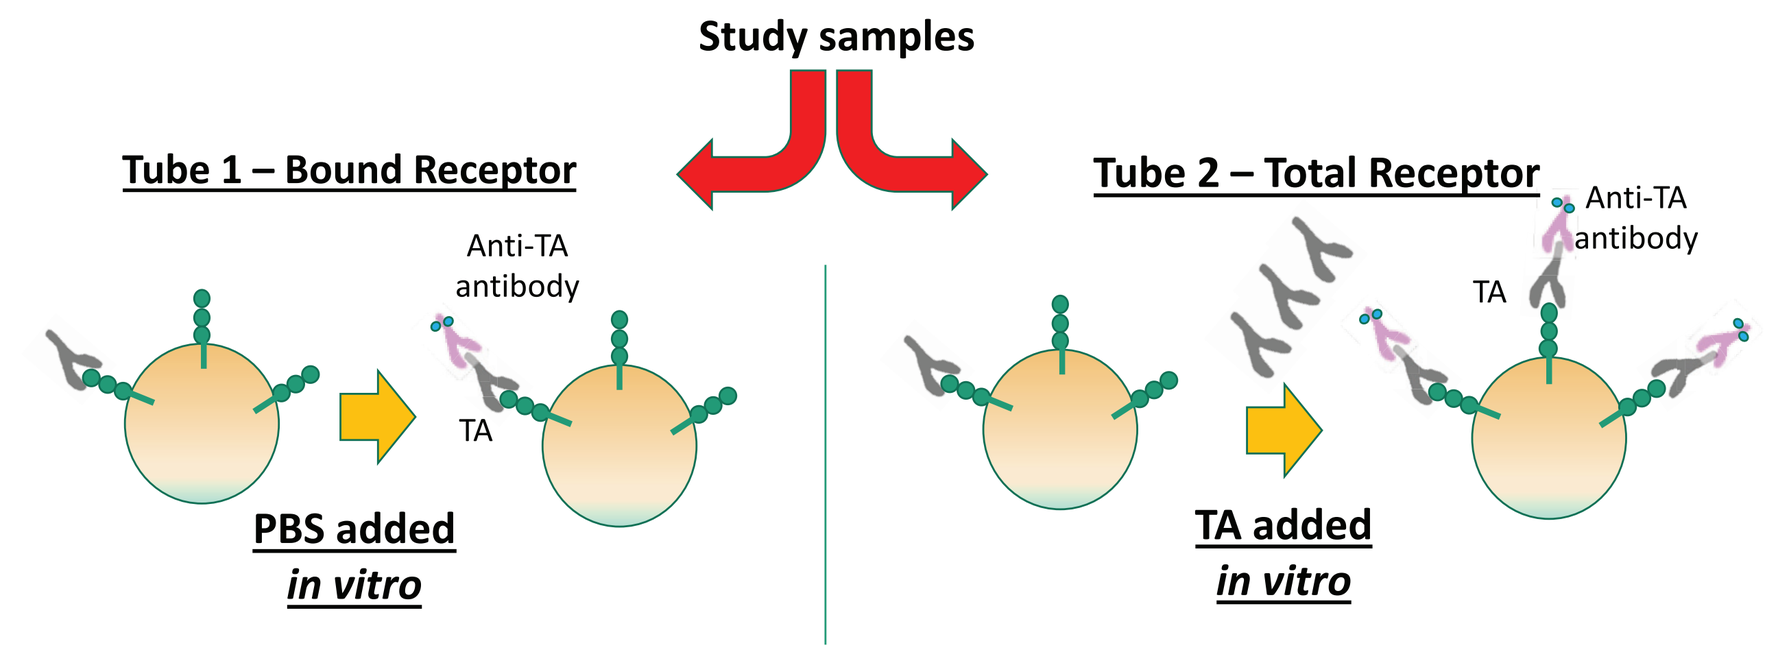
Study samples
Tube 1 – Bound Receptor
Tube 2 – Total Receptor
Anti-TA antibody
TA
PBS added
in vitro
TA added
in vitro

RO assay with secondary antibody to measure Bound and Total Receptor. Study samples from animals that have received TA were split into two tubes. In Tube 1, samples were incubated with PBS *in vitro* followed by staining with secondary antibody to detect Bound Receptor. In Tube 2, samples were incubated with saturating amounts of TA *in vitro* followed by staining with the same secondary antibody to detect Total Receptor.

RESULTS

Figures 3. Assessment of RO by TA to HLA-DR+CD20- antigen presenting cells from whole blood using competing and non-competing antibodies

3a. STEP 1

Titration of non-competing antibody

Non-competing antibody	
Dilution 1	
Dilution 2	← **Saturation**
Dilution 3	
Dilution 4	
Dilution 5	

Titration of competing antibody

Competing antibody	
Dilution 1	
Dilution 2	
Dilution 3	← **Saturation**
Dilution 4	
Dilution 5	

STEP 2 - Multiplexing of competing and non-competing antibodies

Concentration of TA	Non-competing antibody @ Dilution 2	Competing antibody @ Dilution 3	Non-competing + competing antibody
Dilution 1	Tube 1	Tube 2	Tube 3
Dilution 2	Tube 4	Tube 5	Tube 6
Dilution 3	Tube 7	Tube 8	Tube 9
Dilution 4	Tube 10	Tube 11	Tube 12
Dilution 5	Tube 13	Tube 14	Tube 15

Characterization of competing and non-competing antibodies.
Top left: Titration of competing and non-competing antibodies to determine the minimal amount of antibodies needed to saturate entire therapeutic target.
Bottom left: Next, samples were pre-incubated with varying amount of TA, and stained with saturating amount of 1) non-competing antibody only 2) competing antibody only 3) competing and non-competing antibodies. To multiplex competing and non-competing antibodies in one reaction, the staining pattern obtained in the multiplex should be comparable to staining pattern obtained when only one of the two antibodies is used.

3b.

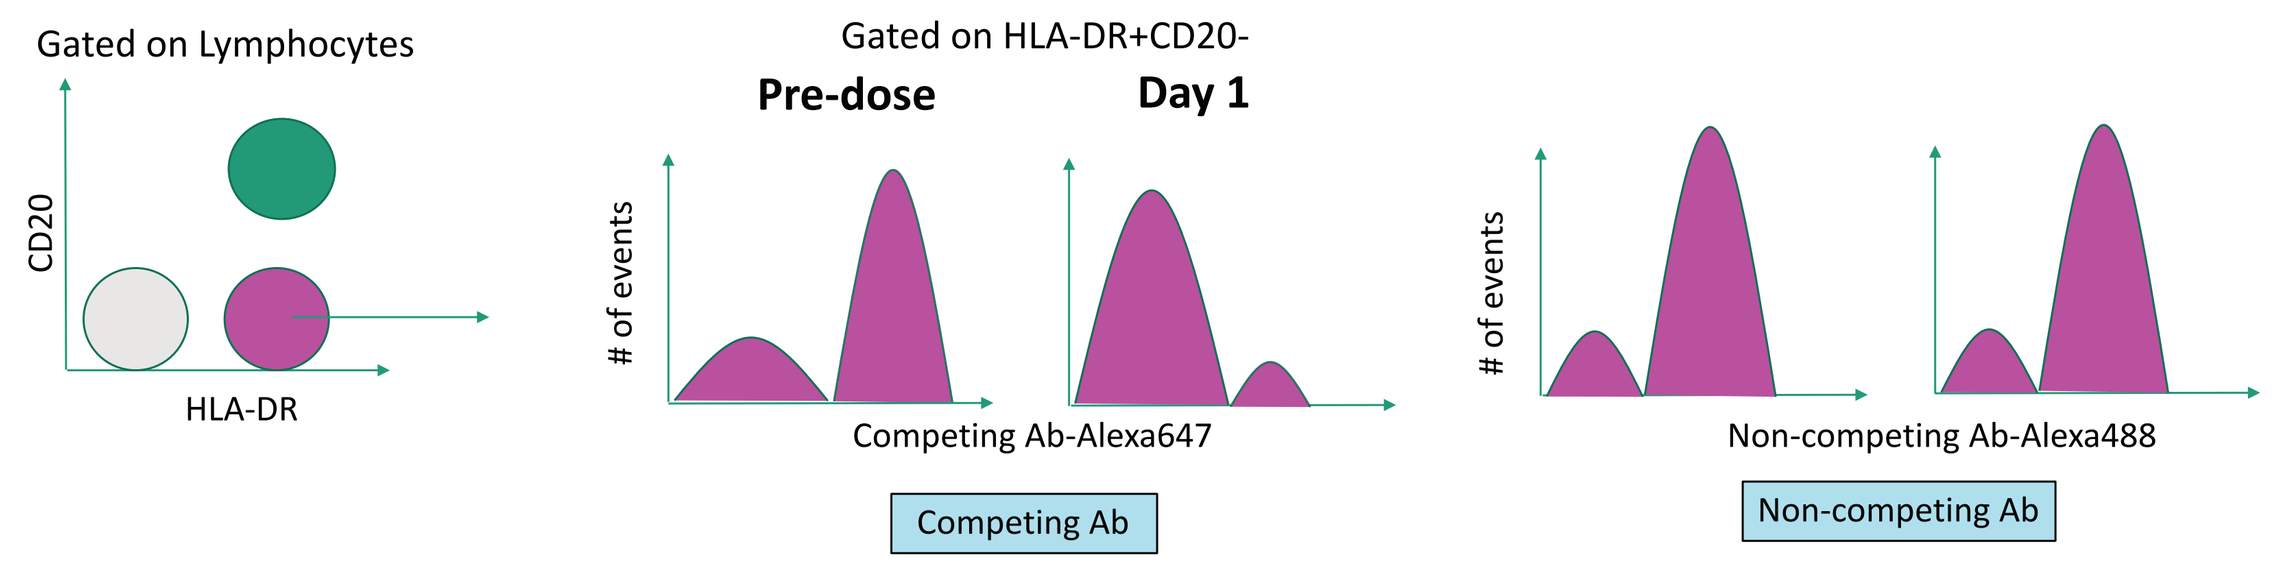
Gated on Lymphocytes
CD20
HLA-DR
Gated on HLA-DR+CD20-
Pre-dose
Day 1
of events
Competing Ab-Alexa647
Competing Ab
Non-competing Ab-Alexa488
Non-competing Ab

Detection of Free and Total receptor with competing and non-competing antibodies, respectively. Whole blood was collected from animals pre-dose and post-dose on Day 1 following TA treatment. Samples were stained with fluorescently labeled antibodies against HLA-DR, CD20, and known TA-competing antibody conjugated to Alexa647 and non-competing antibody conjugated to Alexa488. The therapeutic target was previously identified to be highly expressed on HLA-DR+CD20- antigen- presenting cells and was therefore chosen for RO analysis. Histograms showed lower binding of competing antibody on Day 1 when compared to pre-dose, indicating decreased Free Receptor or available therapeutic target. In contrast, binding of non-competing antibody remained comparable between pre-dose and Day 1.

3c.

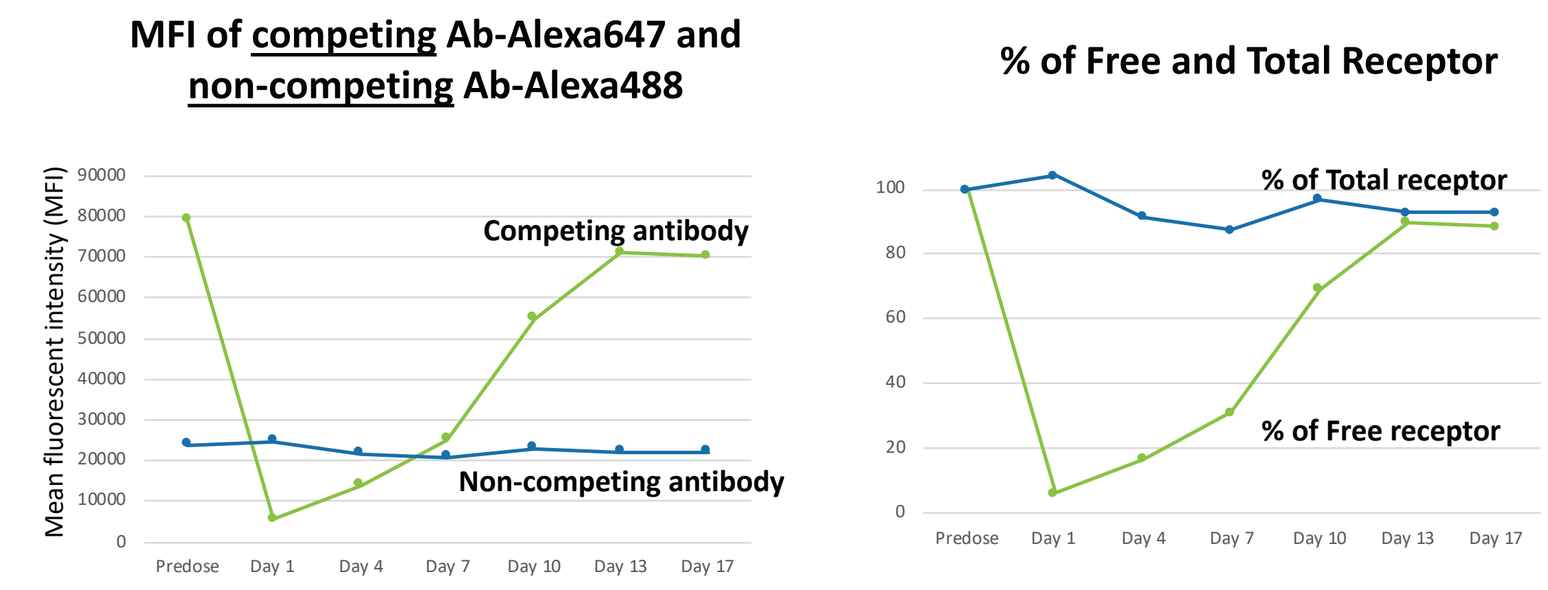
MFI of competing Ab-Alexa647 and non-competing Ab-Alexa488
Mean fluorescent intensity (MFI)
Competing antibody
Non-competing antibody
% of Free and Total Receptor
% of Total receptor
% of Free receptor
Predose
Day 1
Day 4
Day 7
Day 10
Day 13
Day 17

Assessment of Free and Total receptor. Left: Whole blood from animals treated with TA was collected at multiple time points and analyzed using the method described in (A). Mean fluorescent intensity (MFI) of HLA-DR+CD20- cells in the Alexa647 and Alexa488 channels indicated binding of competing and non-competing antibodies, respectively. Binding by non-competing antibody can sometime produce a lower MFI when compared to competing antibody as shown here, likely due to differences in antibody-binding affinity and inherent brightness of the fluorochromes. Right: To adjust for these differences, calculations of percentage of Free and Total Receptor were normalized to pre-dose using the formula listed in Figure 3D.

3d.

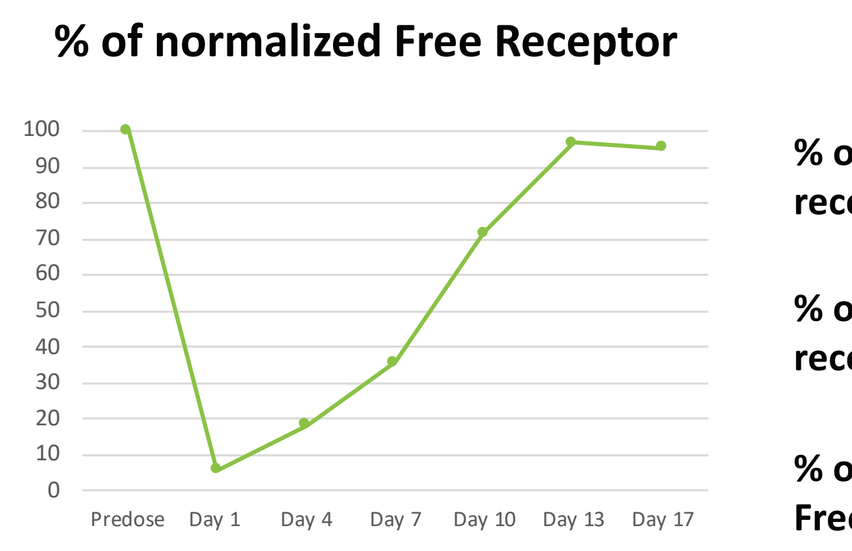
% of normalized Free Receptor
Predose
Day 1
Day 4
Day 7
Day 10
Day 13
Day 17

$$\text{\% of Free receptor} = \frac{MFI\ of\ Competing\ Ab\ on\ Day\ x - FMO}{MFI\ of\ Competing\ Ab\ at\ Predose - FMO} * 100$$

$$\text{\% of Total receptor} = \frac{MFI\ of\ Non\ Competing\ Ab\ on\ Day\ x - FMO}{MFI\ of\ Non\ Competing\ Ab\ at\ Predose - FMO} * 100$$

$$\text{\% of normalized Free receptor} = \frac{\%\ of\ Free\ Receptor\ on\ Day\ x}{\%\ of\ Total\ Receptor\ on\ Day\ x} * 100$$

Normalization of Free Receptor to Total Receptor mitigates effects of receptor modulation. It has been reported that treatment of TA can sometimes result in downregulation of the therapeutic target, leading to lower levels of available target for occupancy by TA. Normalizing the percentage of Free Receptor to the percentage of Bound Receptor will likely produce a more accurate calculation of receptor occupancy of therapeutic target by TA.

Figures 4. Assessment of RO by TA to CD3+ T cells using secondary anti-human IgG antibody

4a.

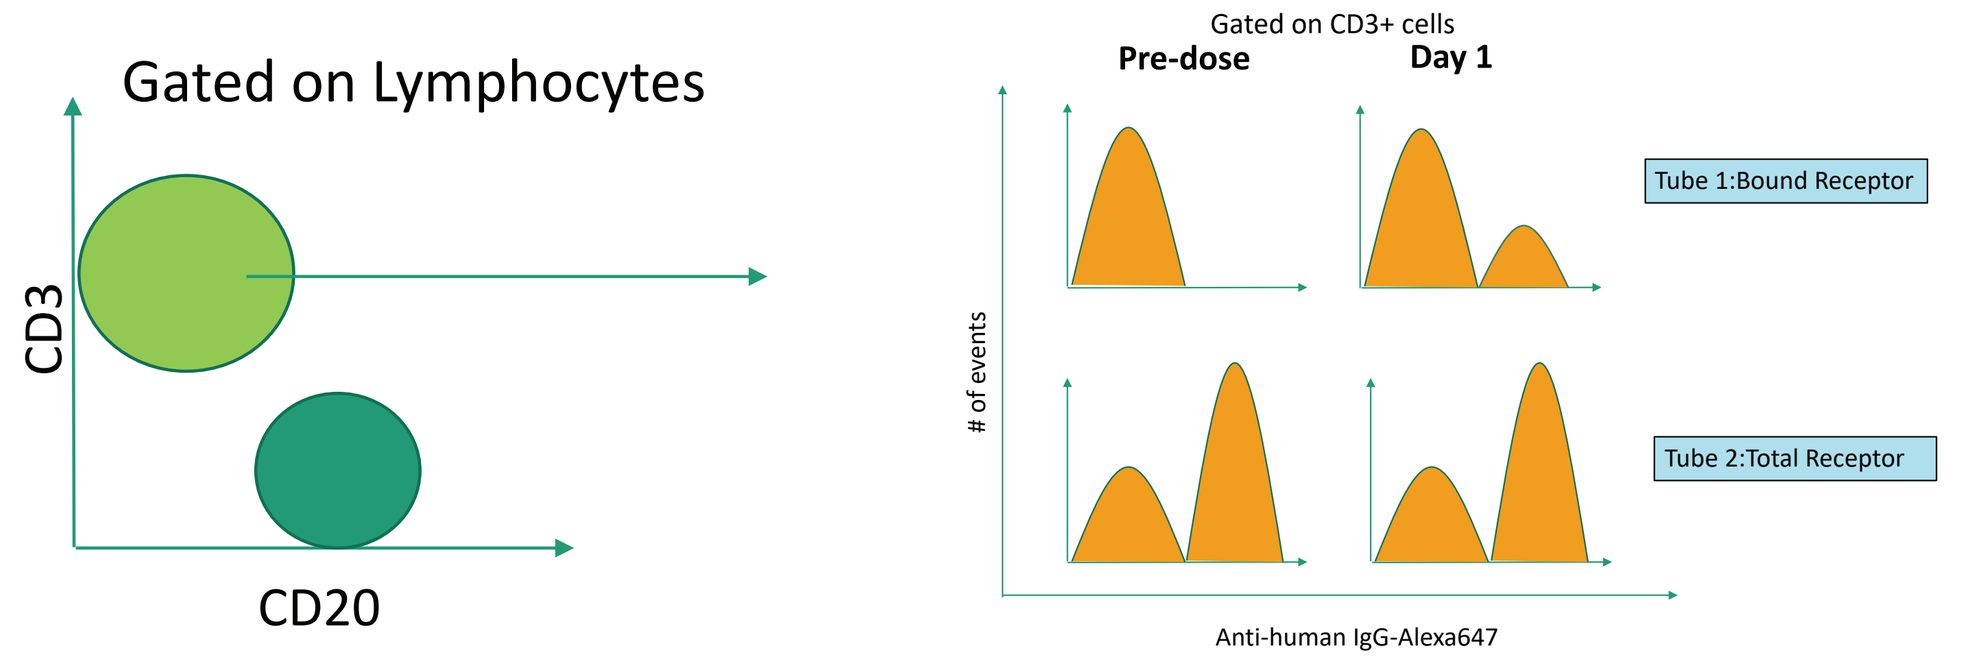
Gated on Lymphocytes
CD3
CD20
Gated on CD3+ cells
Pre-dose
Day 1
of events
Tube 1:Bound Receptor
Tube 2:Total Receptor
Anti-human IgG-Alexa647

Detection of Bound and Total receptor with secondary anti-human IgG. Whole blood was collected from animals pre-dose and post-dose on Day 1 following TA treatment and divided into two tubes for incubation with phosphate buffered saline (PBS, Tube 1) or TA (Tube 2) *in vitro.* Following red blood cell lysis, cells were stained with fluorescently labeled antibodies against CD3, CD20, and anti-human IgG conjugated to Alexa647.

4b.

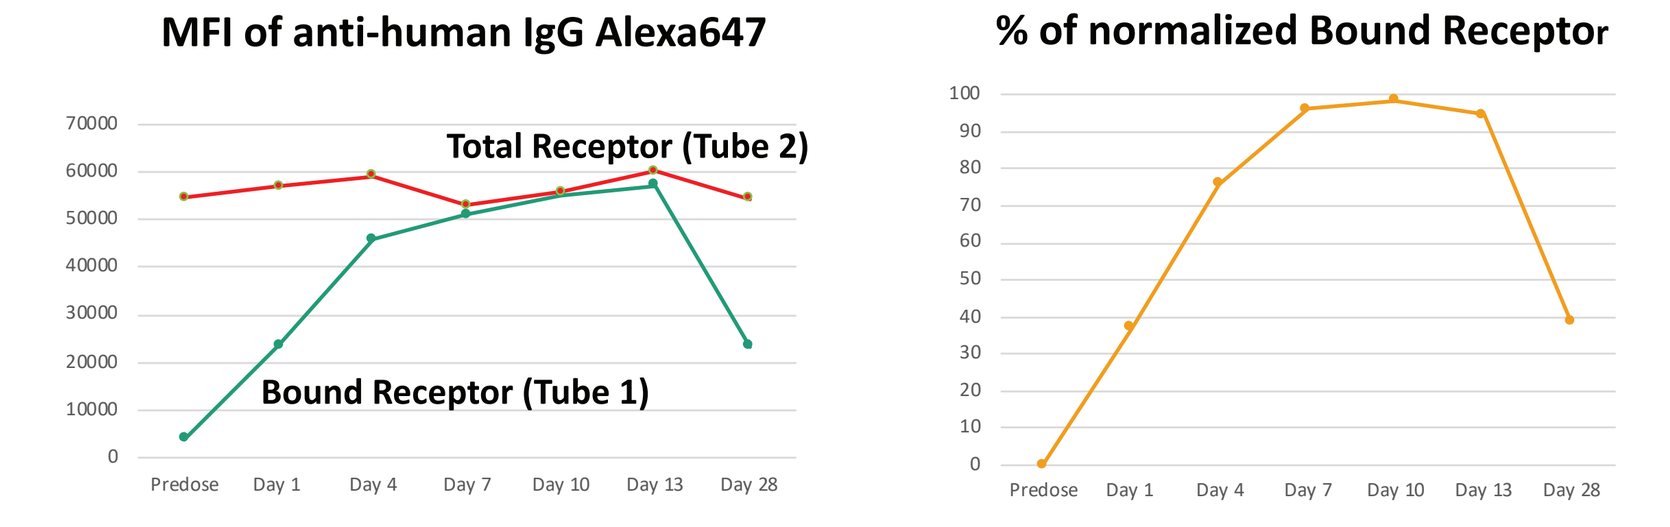
MFI of anti-human IgG Alexa647
Total Receptor (Tube 2)
Bound Receptor (Tube 1)
% of normalized Bound Receptor
Predose
Day 1
Day 4
Day 7
Day 10
Day 13
Day 28

$$\text{\% of normalized Bound Receptor} = \frac{MFI\ of\ Alexa647\ from\ Tube\ 1\ on\ Day\ x - MFI\ of\ Alexa647\ from\ Tube\ 1\ at\ Predose}{MFI\ of\ Alexa647\ from\ Tube\ 2\ on\ Day\ x - MFI\ of\ Alexa647\ from\ Tube\ 1\ at\ Predose} * 100$$

Assessment of Bound and Total receptor and calculating percentage of normalized Bound Receptor. **Left:** Whole blood from animals treated with TA was collected at multiple time points and analyzed using the method described in (A). Mean fluorescent intensity (MFI) of CD3+ T cells in the Alexa647 channel indicated binding of the secondary anti-human IgG antibody to TA on therapeutic target (Bound Receptor, Tube 1) and the total available therapeutic target expressed on target cells (Total Receptor, Tube 2). **Right:** Normalization of Bound Receptor to Total Receptor will likely produce a more accurate calculation of receptor occupancy of therapeutic target by TA. The formula used to calculate percentage of normalized Bound Receptor also subtracts non-specific binding observed in the absence of TA (Tube 1 pre-dose).

Figures 5. Comparison of two secondary anti-human IgG (anti-TA) antibodies

5a.

Features	Antibody #1	Antibody #2
Product name	AffiniPure F(ab')$_2$ Fragment Goat Anti-Human IgG, Fcγ fragment specific	Goat Anti-Human IgG, Monkey absorbed
Manufacturer	Jackson Immunoresearch	Southern Biotech
Catalogue #	109-136-098	2049-31
Pre-absorption	**Bovine, Horse, Mouse Serum Proteins**	**Human IgM and IgA; rhesus (Macaca mulatta) and cynomolgus (Macaca fascicularis) IgG**
Clonality	Polyclonal	Polyclonal
Isotype	Goat IgG	Goat IgG

Selection of an appropriate anti-human Ig antibody for detection of TA can have a significant impact on the results. When choosing between comparable secondary antibodies, one feature that should be considered is the pre-absorption step performed by the manufacturer to minimize cross-reactivity to other species.

5b.

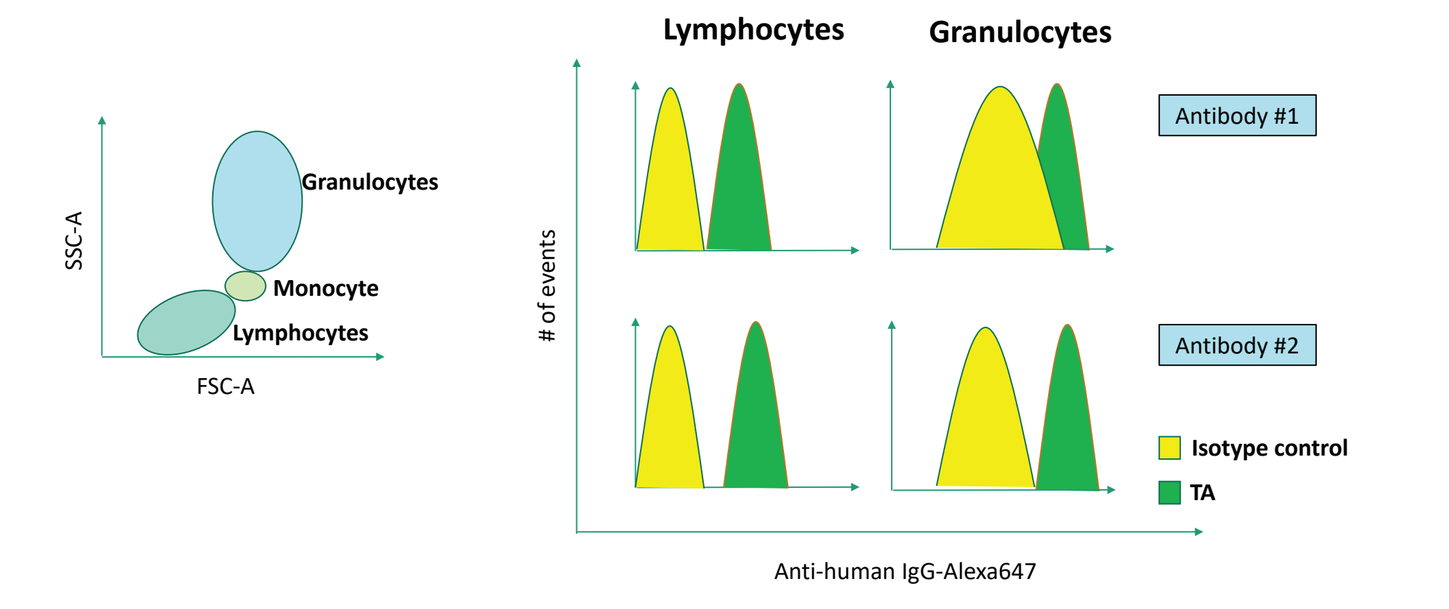
SSC-A
FSC-A
Granulocytes
Monocyte
Lymphocytes
Granulocytes
of events
Antibody #1
Antibody #2
Isotype control
TA
Anti-human IgG-Alexa647

Secondary antibody affects resolution of cell population. For cell types with low expression of Fc receptors such as T cells, the choice of secondary antibody can still have a minor impact on the resolution between positive and negative populations. For cell types such as monocytes and granulocytes, the secondary antibody can have a major impact on the resolution signal and noise.

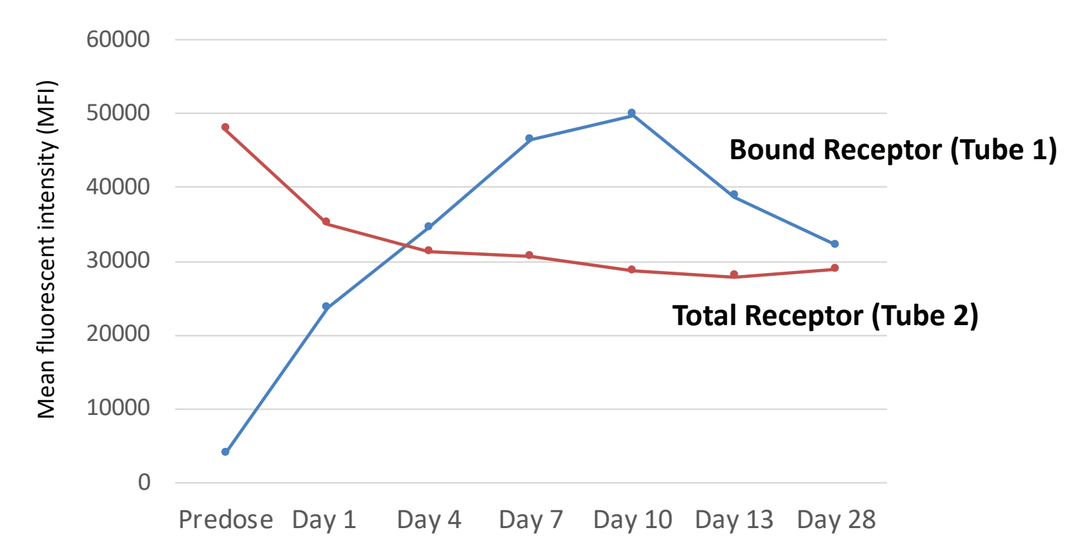
Mean fluorescent intensity (MFI)
Bound Receptor (Tube 1)
Total Receptor (Tube 2)
Predose
Day 1
Day 4
Day 7
Day 10
Day 13
Day 28

Figures 6. Receptor modulation following re-exposure to TA *in vitro*
RO assessment using secondary antibody to assess Total Receptor expression by saturating all therapeutic targets with TA *in vitro* can sometimes lead to receptor modulation. This was evident when samples collected after dosing in vivo exhibited lower levels of Total Receptor when compared to samples collected from the same animal prior to dosing. This observation suggested that some therapeutic targets will undergo receptor modulation when samples are re-exposed to TA *in vitro*.

CONCLUSION

Here, we discussed two methods to assess RO via flow cytometry and present the benefits and pitfalls of both methods. Assessment of RO using competing and non-competing antibodies to measure Free and Total Receptor enables multiplexing in one tube when sample volume is limited, but identification and characterization of both antibodies can be laborious and time consuming. In contrast, a RO assay using secondary anti-human IgG antibody can be set up quickly, but will require more samples for separate assessment of Bound and Total Receptor. In addition, identification of the appropriate secondary and blocking reagents are crucial and receptor downregulation may occur following re-exposure of TA *in vitro*. The technical considerations presented here, when incorporated early in the RO design phase, will aid in determining the most suitable approach.

Reference: https://www.ncbi.nlm.nih.gov/pubmed/26054054

© 2020 Altasciences. All Rights Reserved.